MEDICAL UNIVERSITY
OF VIENNA

Volumetric DCE-MRI Analysis for Breast Cancer Detection

Barbara Bennani-Baiti, MD, PhD, MS, Thomas Helbich MD, MS, Michael Weber, PhD, MS, Pascal Baltzer, MD

Universitätsklinik für Radiologie und Nuklearmedizin, Medizinische Universität Wien

Objective

While computer aided detection (CAD) is already routinely employed in conventional mammography imaging, few data are available on CAD cancer detection at MRI[1,2]. Given its excellent diagnostic performance (sensitivities >95%), breast MRI is being increasingly used in routine imaging. An automated approach pre-assessing the risk of malignancy would be most helpful in assisting inexperienced readers[3-5]. We therefore aimed to assess whether CAD based on volumetric DCE-MRI analysis could predict the presence of malignancy.

Patients and Methods

This retrospective IRB-approved study, included 39 consecutive patients, that underwent two subsequent breast MRI exams for suspicious findings at conventional imaging with 0.1 mmol/kg gadobenic and gadoteric acid. Two independent readers, blinded to the histopathological outcome, assessed unenhanced and early post-contrast images using computer-assisted software (Brevis, Siemens Healthcare)[6]. Diagnostic performance was statistically determined for percentage of ipsilateral voxel volume enhancement (see Table 1 and Figures 1 and 2) after crosstabulation with the dichotomized histological outcome (benign/malignant).

imaging biomarker	definition
early enhancement	$\frac{\textit{total number of all threshold-passing enhancing voxels within VOI}}{\textit{all voxels within VOI}}$

Table 1. Tabellary view of employed method.

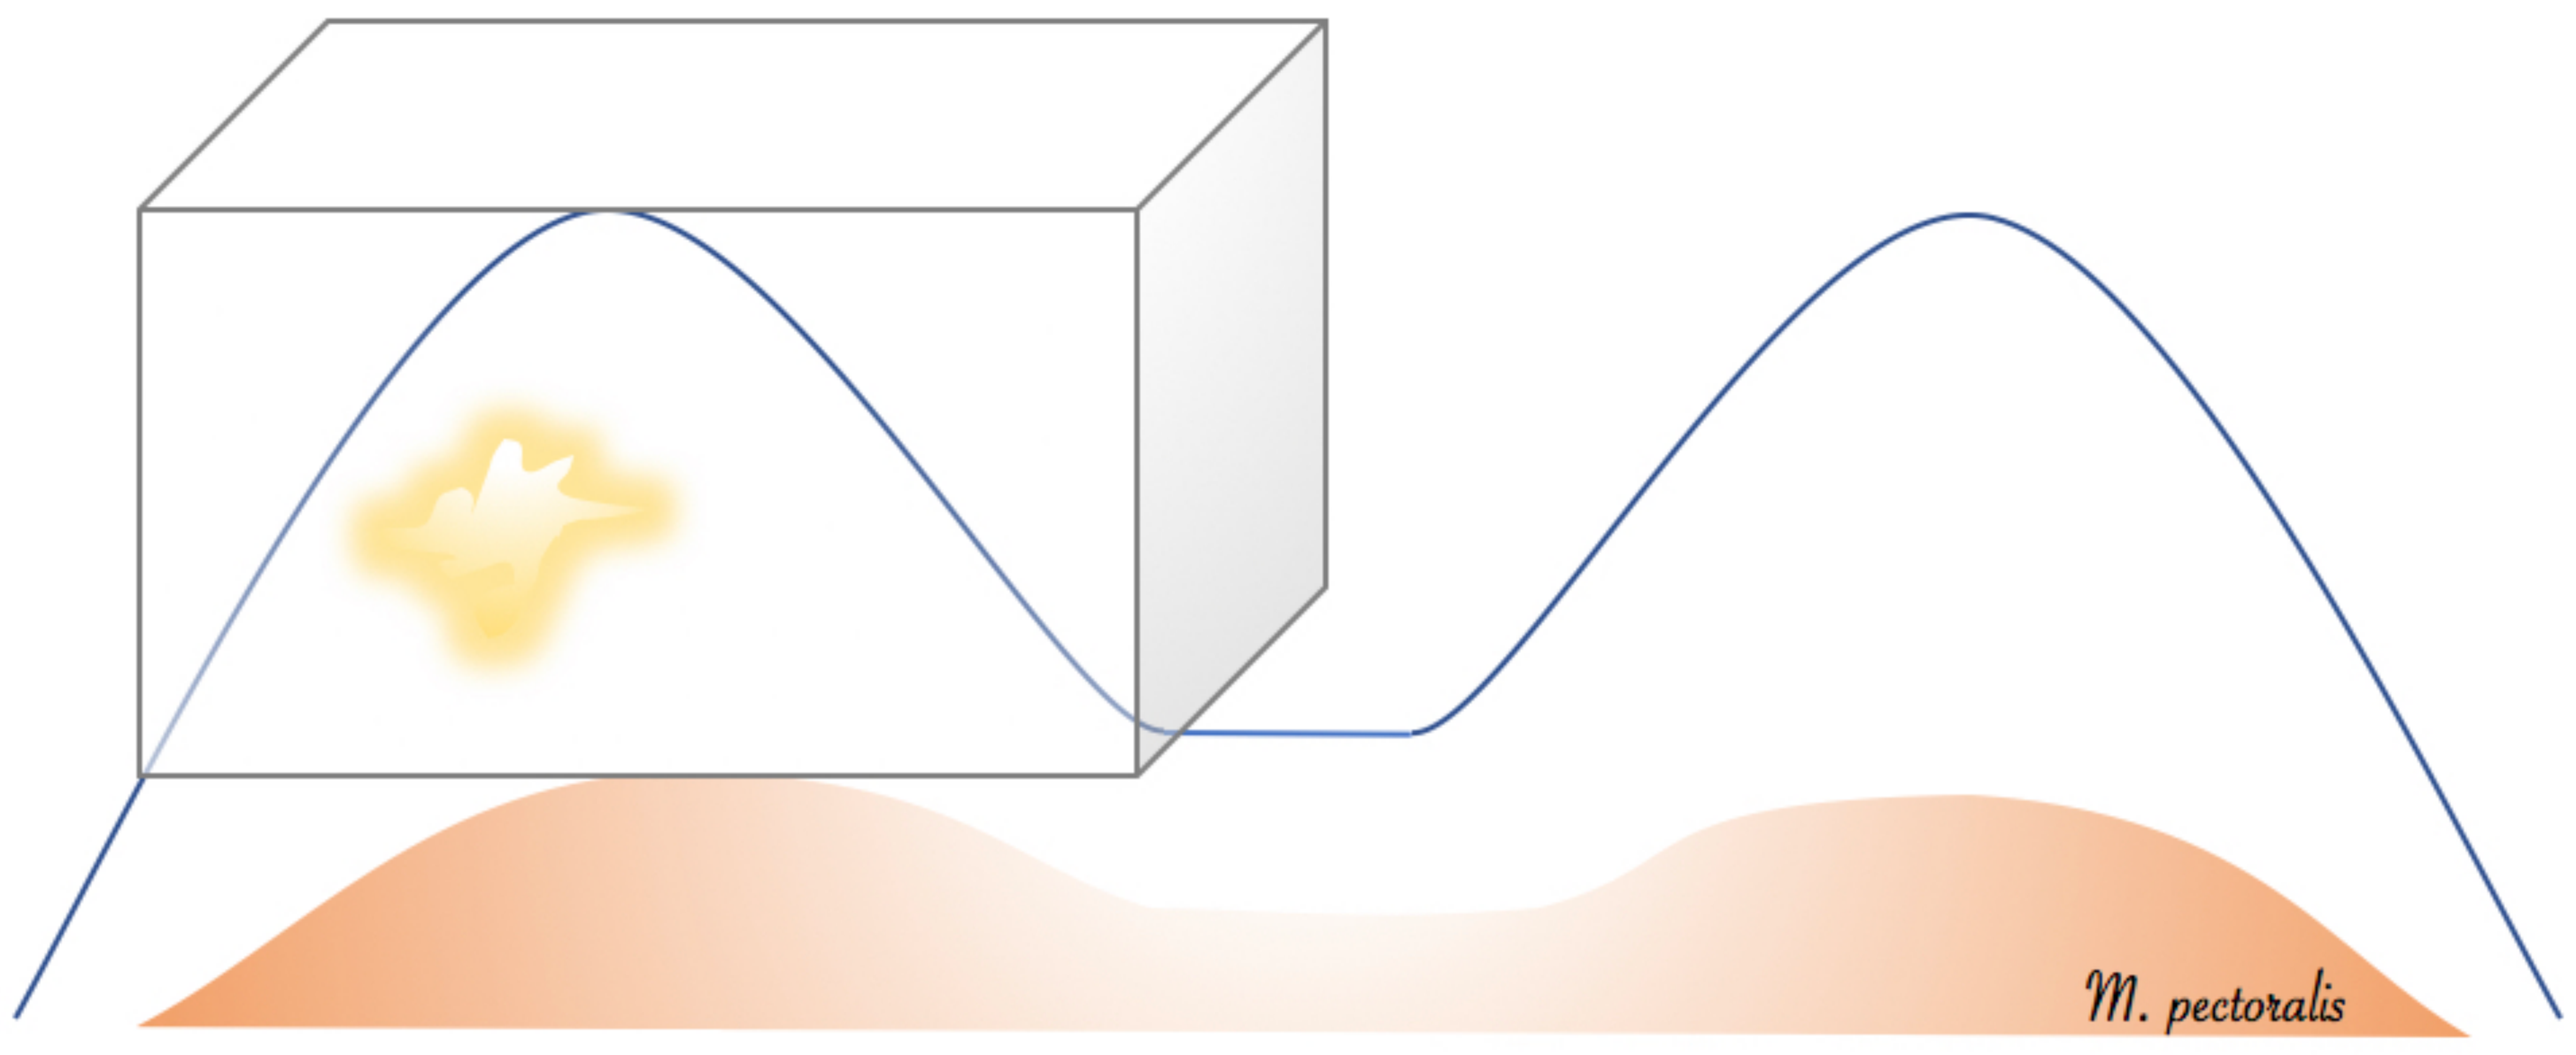
M. pectoralis

Figure 1. Schematic drawing of VOI placement for enhancing voxel assessment. The cubic VOI spans the entire breast and ends shorty before the M. pectoralis.

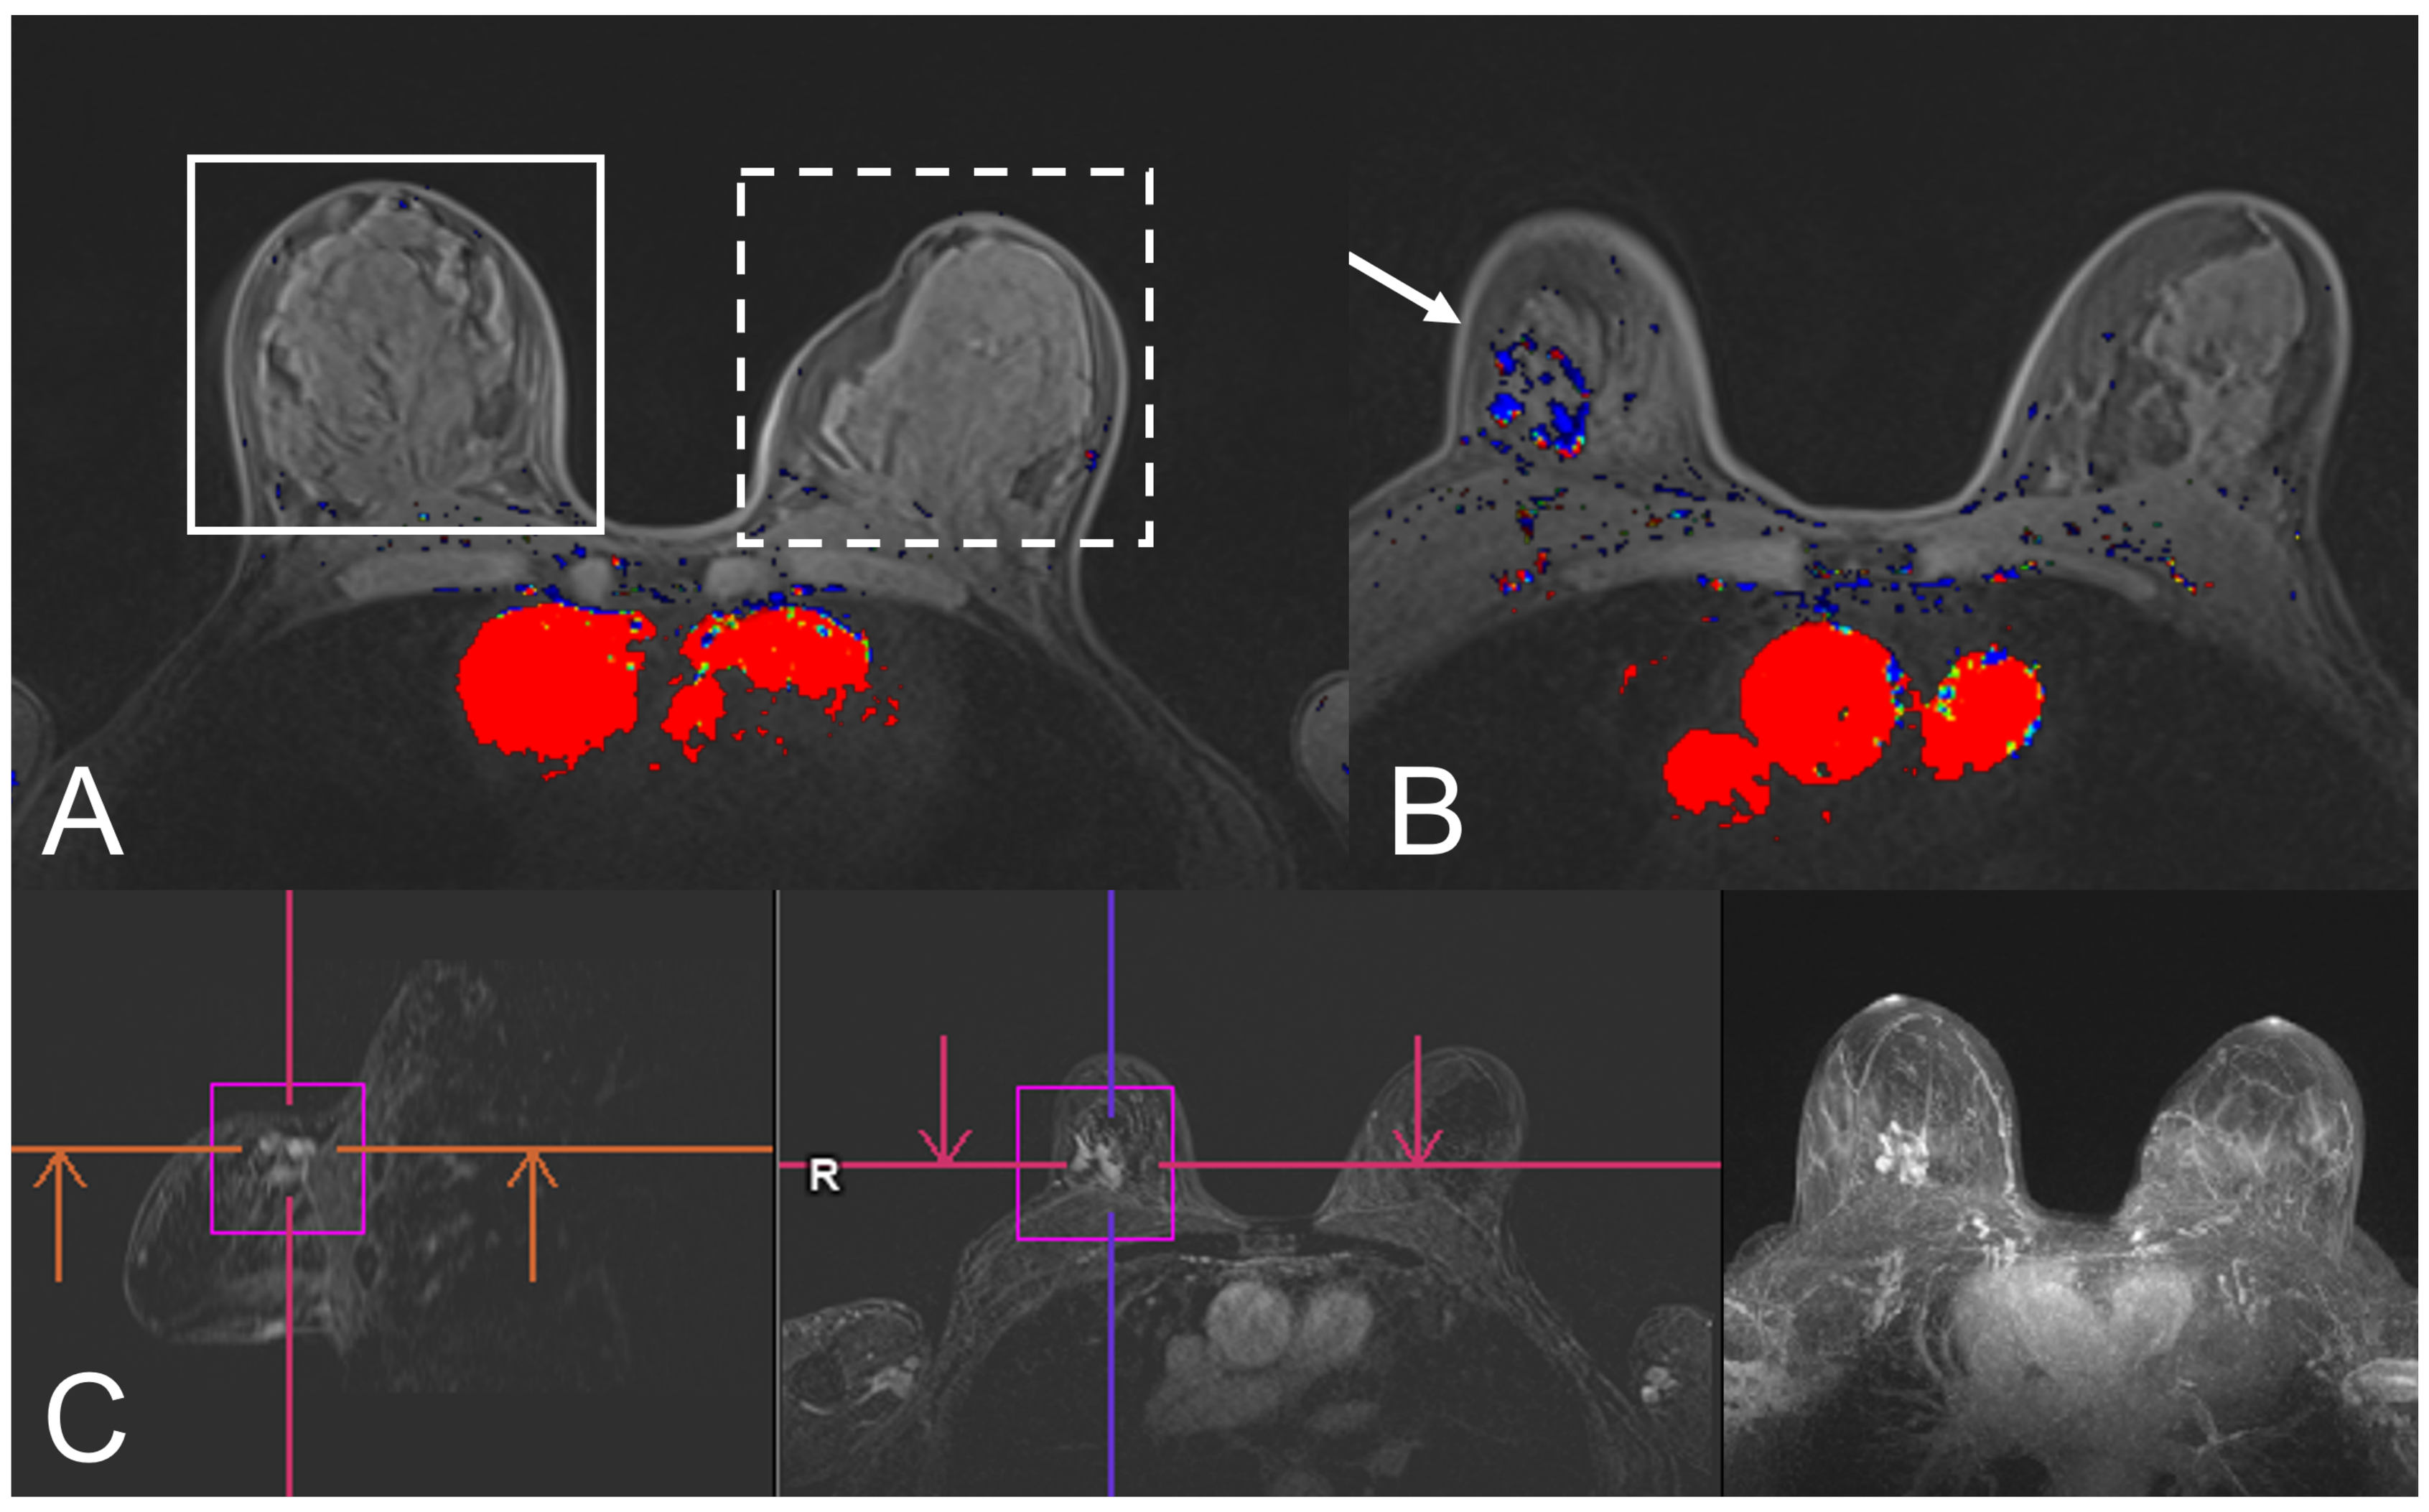
A
B
C
R

Figure 2: Enhancing voxel assessment using semiautomated software (BreVis, Siemens Healthineers, Erlangen Germany). Exemplary case of a postmenopausal woman with luminal B type cancer of the right breast. (A) A user-defined cubic Volume of Interest (VOI) is placed around the entire breast. The solid and dashed lines delineate the VOIs corresponding to the "ipsilateral" and "contralateral" sides, respectively. The software calculates enhancing voxel volume and distribution of initial and delayed enhancement curve types. A threshold of 20% was set to account for noise. Only threshold-passing voxels were accounted for in the volumetric analysis. (B) enhancement corresponds to the malignancy positioned cranially in the right breast (C) multiplanar reconstruction.

Results

39 lesions (33 malignant, 6 benign) were assessed. For patient and histopathological characteristics see Table 2. Ipsilateral enhancing voxel volume versus histological outcome indicated an AUC of 0.707 and 0.687 for gadobenic acid, reader 1 and 2, respectively and an AUC of 0.778 and 0.773 for gadoteric acid, reader 1 and 2, respectively (Figure 3). Excellent interreader agreement was achieved for both measurements, with an intraclass correlation (ICC) of 0.98 (95%-CI 0.97-0.99) and 0.99 (0.98-0.99) for gadobenic acid and gadoteric acid, respectively (Figure 3). Pairwise testing yielded no statistically significant difference both between readers and between contrast agents employed (p>0.05).

Characteristic	n (%)
Entire cohort	39 (100%)
Mean patient age (SD)	57.5 (±12.1y)
Tumor grade	
G1	6 (18.2%)
G2	16 (48.5%)
G3	11 (33.3%)
benign	6 (15.4%)
Mean patient age (SD)	54.2 (±17.6y)
Lesion (B2)	4 (33.3%)
Inflammation	2 (66.6%)
malignant	33 (84.6%)
Mean patient age (SD)	58.2 (±11.1y)
Luminal A	7 (21.2%)
Luminal B	15 (45.5%)
HER2-positive	6 (18.2%)
TN/basal-like	5 (15.2%)

Table 2. Clinical and histopathological properties of participants (n = 39). Abbreviations: G1, grade 1; G2, grade 2; G3, grade 3; HER2; human epidermal growth receptor 2; SD, standard deviation; TN, triple negative.

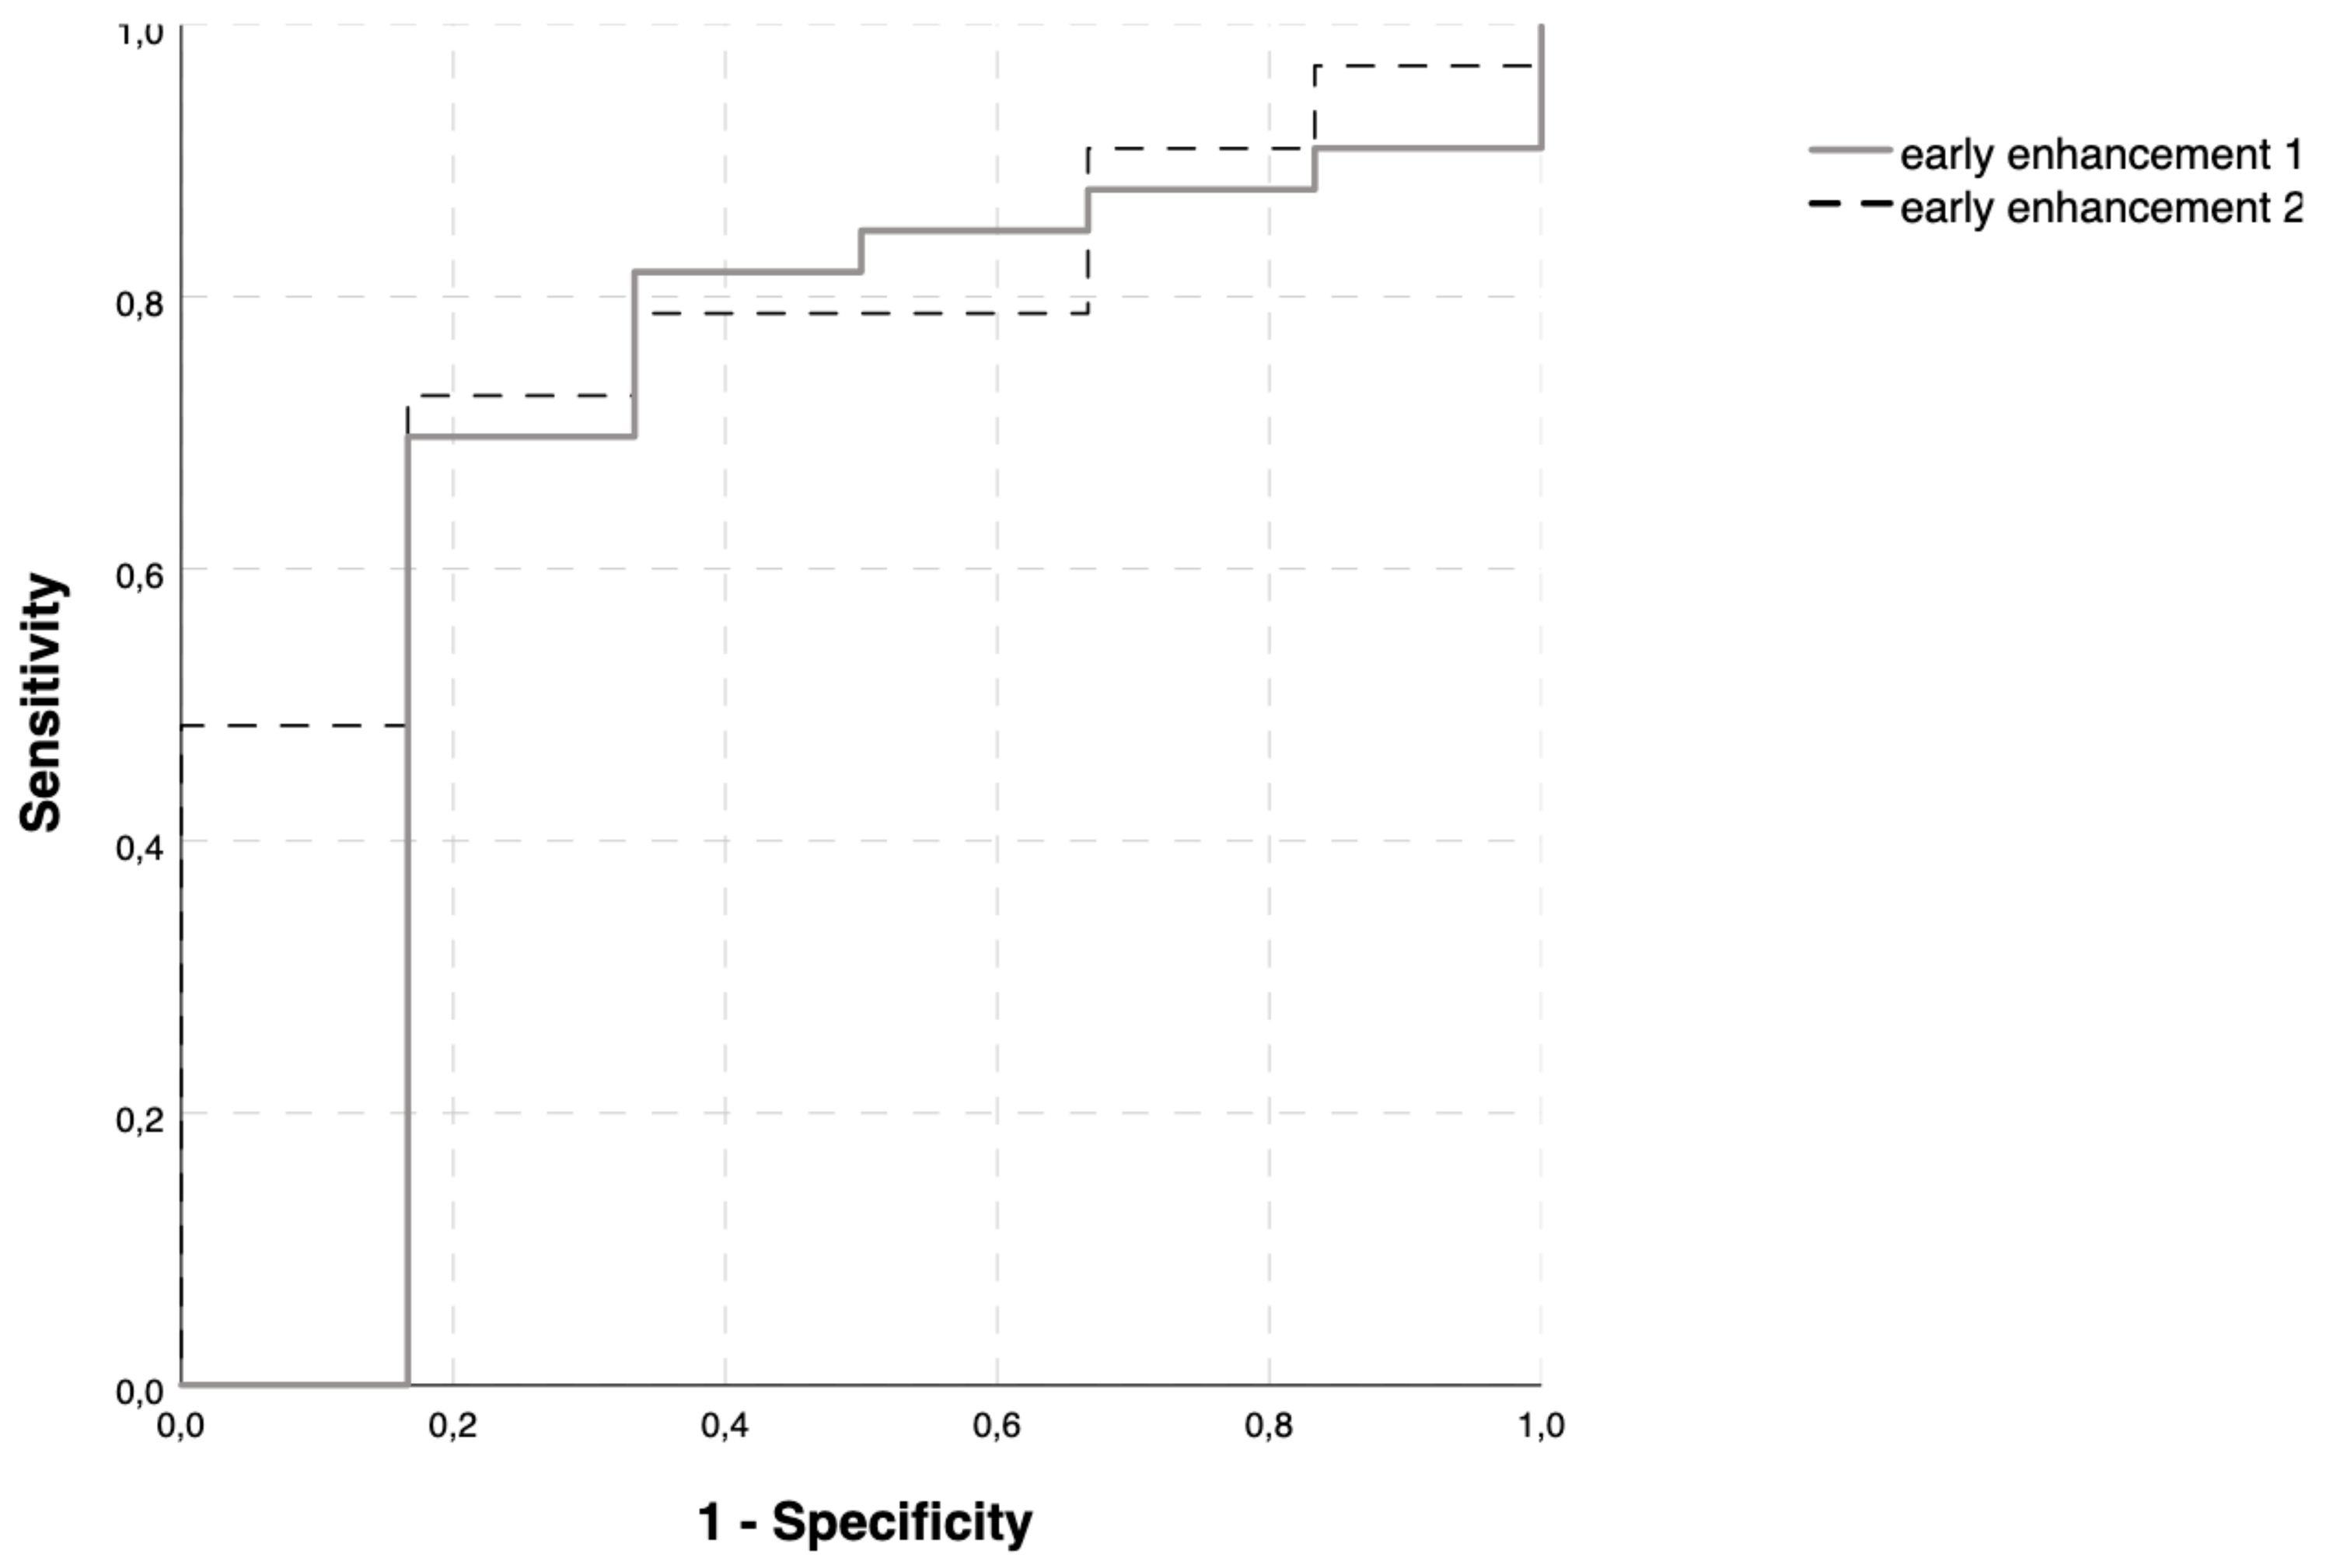
Sensitivity
1 - Specificity
0,0
0,2
0,4
0,6
0,8
1,0
early enhancement 1
early enhancement 2

Figure 3: ROC curves. Early enhancement 1 and 2 refer to percentage of early enhancement utilizing gadobenic and gadoteric acid, respectively.

Conclusion

The findings from this pilot study suggest that quantitatively assessed enhancing breast tissue as a percentage of the entire breast volume may serve as an indicator for breast cancer. However, differences in contrast media should be accounted for, before further developing this method.

Ackknowledgement

Study data were acquired within the context of a propsective randomized diagnostic study sponsored by BRACCO (Milan, Italy).

References

1. Song SE, Seo BK, Cho KR, et al. Computer-aided detection (CAD) system for breast MRI in assessment of local tumor extent, nodal status, and multifocality of invasive breast cancers: preliminary study. Cancer Imaging. 2015;15:1.
2. Renz DM, Böttcher J, Diekmann F, et al. Detection and classification of contrast-enhancing masses by a fully automatic computer-assisted diagnosis system for breast MRI. J Magn Reson Imaging. 2012;35(5):1077–1088.
3. Bennani-Baiti B, Bennani-Baiti N, Baltzer PA. Diagnostic Performance of Breast Magnetic Resonance Imaging in Non-Calcified Equivocal Breast Findings: Results from a Systematic Review and Meta-Analysis. PLoS ONE. 2016;11(8):e0160346.
4. Houssami N, Ciatto S, Macaskill P, et al. Accuracy and surgical impact of magnetic resonance imaging in breast cancer staging: systematic review and meta-analysis in detection of multifocal and multicentric cancer. J Clin Oncol. 2008;26(19):3248–3258.
5. Warner E, Messersmith H, Causer P, Eisen A, Shumak R, Plewes D. Systematic review: using magnetic resonance imaging to screen women at high risk for breast cancer. Ann Intern Med. 2008;148(9):671–679.
6. Baltzer PAT, Renz DM, Kullnig PE, Gajda M, Camara O, Kaiser WA. Application of computer-aided diagnosis (CAD) in MR-mammography (MRM): do we really need whole lesion time curve distribution analysis? Acad Radiol. 2009;16(4):435–442.